Safety and Efficacy of RT234 Vardenafil Inhalation Powder on Exercise Parameters in Pulmonary Arterial Hypertension: Phase 2b, Dose-Escalation Study Design

Raymond L. Benza,[1] Veronica Franco,[1] Mandar A. Aras,[2] Leslie Spikes,[3] Daniel Grinnan,[4] Carol Satler[5]

[1]The Ohio State University Wexner Medical Center, Columbus, OH, USA; [2]University of California San Francisco, San Francisco, CA, USA; [3]University of Kansas Medical Center, Kansas City, KS, USA; [4]Virginia Commonwealth University School of Medicine, Richmond, VA, USA; [5]Respira Therapeutics, Inc., Boston, MA, USA

BACKGROUND

- Pulmonary arterial hypertension (PAH; Group 1 pulmonary hypertension) is a rare disease defined by abnormally high mean pulmonary arterial pressure (≥20 mmHg)[1]
- PAH is currently managed using chronic, scheduled treatments to improve exercise capacity and delay clinical worsening
- Availability of an as-needed (PRN) treatment for rapid symptom resolution would further enhance patient quality of life
- RT234 is an investigational drug/device combination of vardenafil hydrochloride and the novel Axial Oscillating Sphere (AOS™) dry powder inhaler

OBJECTIVE

- RT234 CL202 is a multicenter, open-label, dose-escalation, Phase 2b trial (NCT04266197) that will evaluate the efficacy and safety of RT234 as assessed by changes in peak oxygen consumption (VO_2) by cardiopulmonary exercise testing (CPET) and the 6-minute walk test (6MWT)

Figure 1: The AOS DPI device used in RT234 (Respira Therapeutics, Inc., Albuquerque, NM, USA)

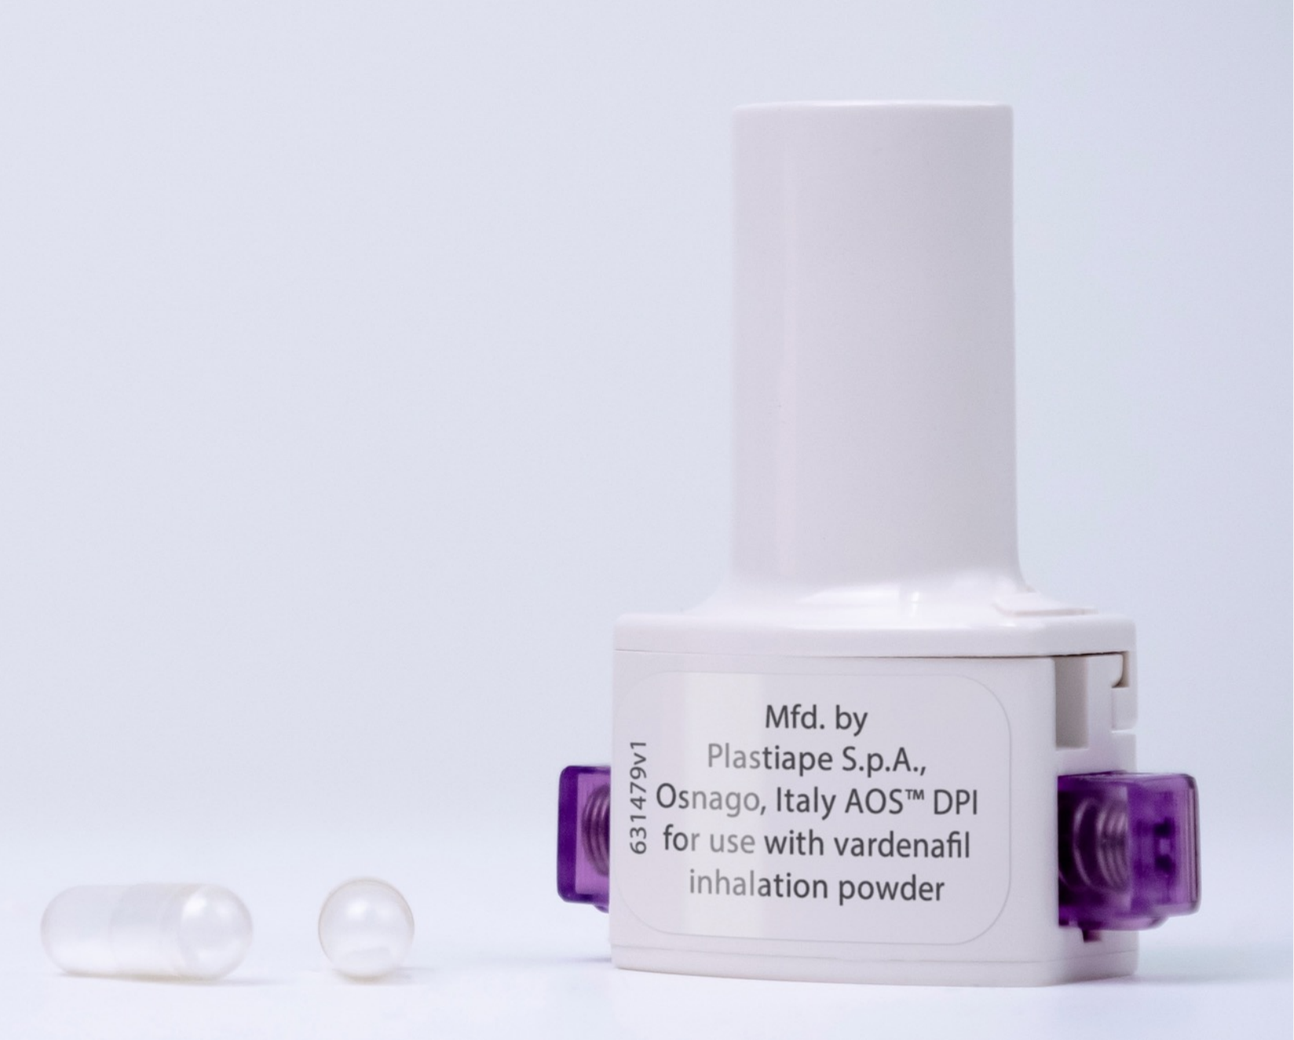
Mfd. by
Plastiape S.p.A.,
Osnago, Italy AOS™ DPI
for use with vardenafil
inhalation powder
631479v1

AOS, Axial Oscillating Sphere; DPI, dry powder inhaler.

METHODS

Figure 2: Study design, CL202

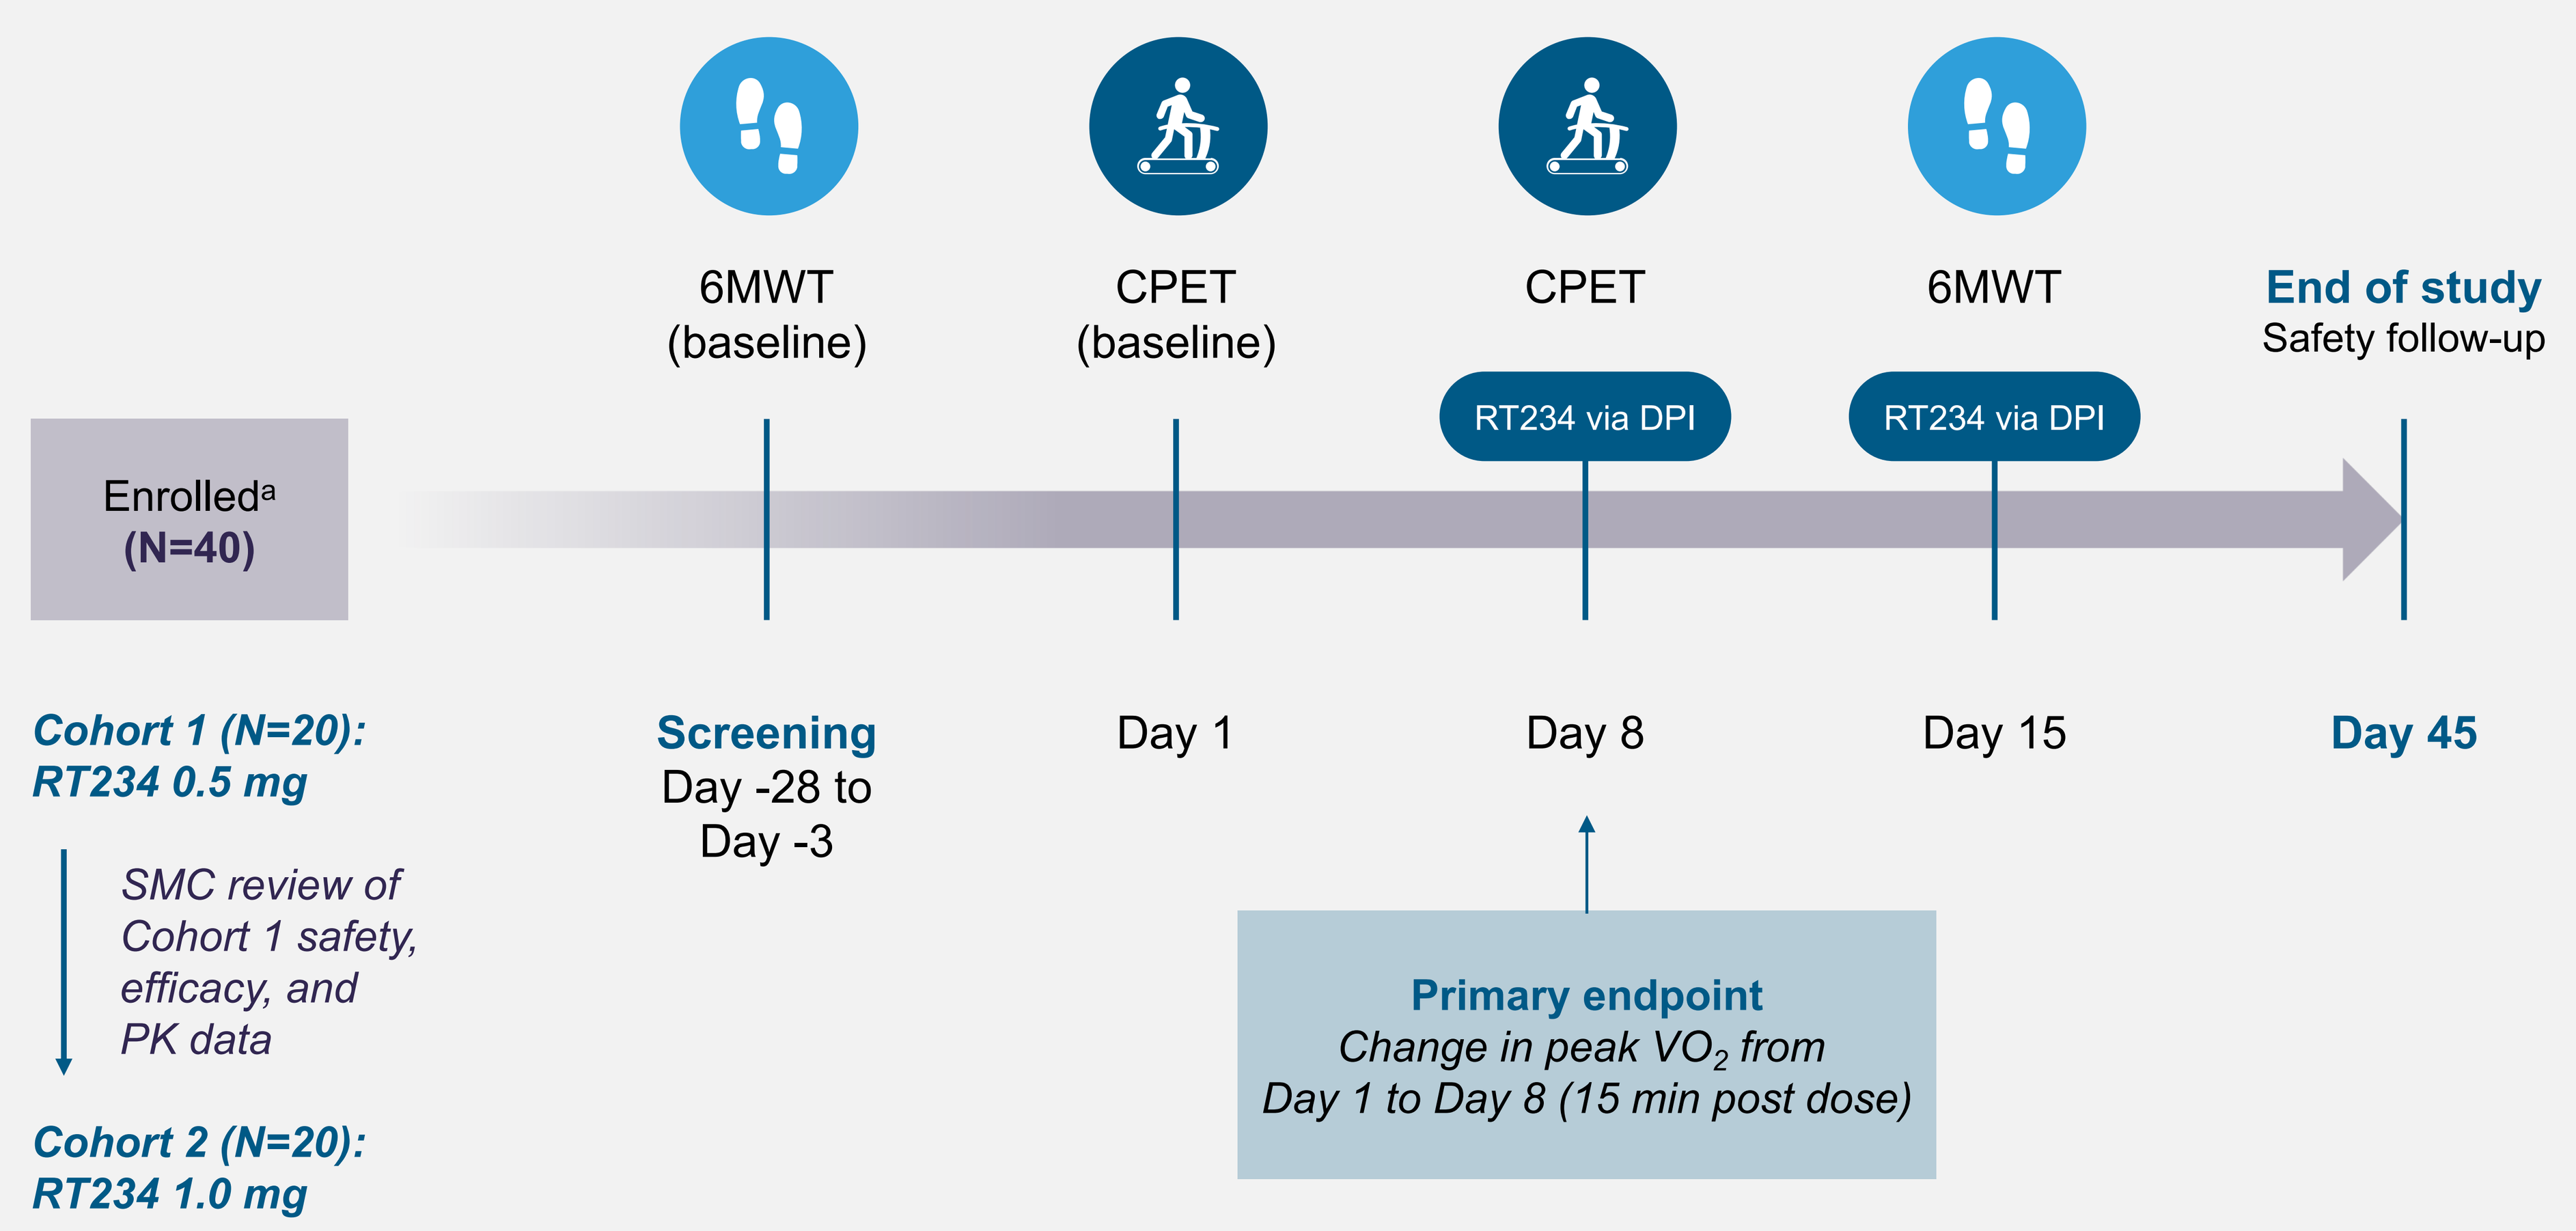
Prospective, multicenter, open-label, 2-cohort, dose-escalation, Phase 2b study
6MWT (baseline)
CPET (baseline)
CPET
6MWT
End of study
Safety follow-up
RT234 via DPI
RT234 via DPI
Enrolled[a] (N=40)
Cohort 1 (N=20): RT234 0.5 mg
SMC review of Cohort 1 safety, efficacy, and PK data
Cohort 2 (N=20): RT234 1.0 mg
Screening
Day -28 to Day -3
Day 1
Day 8
Day 15
Day 45
Primary endpoint
Change in peak VO2 from Day 1 to Day 8 (15 min post dose)

[a] Patients with PAH on stable, disease-specific PAH background therapy with limited exercise capacity.

6MWT, 6-minute walk test; CPET, cardiopulmonary exercise testing; DPI, dry powder inhaler; PAH, pulmonary arterial hypertension; PK, pharmacokinetic; SMC, Safety Monitoring Committee; VO_2, oxygen consumption.

Statistical analyses

- The planned sample size is 40 patients, with a 20% dropout adjustment
 - The estimated power to detect a change in peak VO_2 of 1.5 mL O_2 $kg^{-1}min^{-1}$ between baseline and post RT234 is ≥80%
- The primary endpoint will be analyzed by 1-sample t-test (if peak VO_2 follows a normal distribution) or 1-sample Wilcoxon signed-rank test (if the normality assumption is rejected)
- Hochberg step-up procedure will be used to account for multiplicity of secondary endpoint analyses
- Safety data will be summarized descriptively

Key eligibility criteria

Inclusion

- Aged 18–80 years, inclusive
- Right heart catheterization–confirmed PAH
- On ≤2 stable oral and/or inhaled PAH-specific therapies
- 6-minute walk distance (6MWD) ≥50 m
- CPET with minute ventilation/volume of exhaled CO_2 (VE/VCO_2) slope ≥36

Exclusion

- Baseline systemic hypotension (mean arterial pressure <50 mmHg or systolic blood pressure [SBP] <90 mmHg)
- Uncontrolled hypertension (SBP >175 mmHg or sitting diastolic blood pressure >110 mmHg)
- On parenteral PAH medication or riociguat
- Current or previous left-sided heart disease and/or clinically significant cardiac disease

Key assessments

Primary efficacy endpoint

- Change in peak VO_2 during CPET

Secondary efficacy endpoints

Effect on exercise capacity

- Change in 6MWD measured during a 6MWT
- Change in VE/VCO_2 slope
- Change in end-tidal CO_2 apex response to exercise (highest level during CPET)
- Change in duration of exercise during CPET

Effect on exertional symptoms of PAH

- Change in peak perceived dyspnea during CPET, assessed by Modified Borg Dyspnea Scale Score
- Change in Patient Global Impression of Severity (PGI-S) for CPET
- Change in PGI-S for 6MWT

Exploratory endpoints

- Pharmacokinetics
- Exposure–response analysis

Safety

- Treatment-emergent adverse events
- Vital signs
- Physical examination
- 12-lead electrocardiogram

Enrollment

- Enrollment began in September 2020 and the trial is expected to be completed in December 2023
- The trial will be conducted at 25–30 sites in the United States

CONCLUSIONS

- RT234 is a first-in-class treatment for exertional symptoms of PAH, addressing the unmet clinical need for a PRN treatment for this disease
- If the CL202 trial is successful, it is anticipated that RT234 PRN therapy will provide acute improvements in VO_2 in patients with PAH
- The results of this trial are expected to inform the design of Phase 3 trials of RT234

Please scan code to view poster or visit https://bit.ly/3LjrMIQ

REFERENCE

1. Simonneau G, et al. *Eur Respir J.* 2019;53:1801913.

DISCLOSURES

- R. L. Benza: Steering committee member for Abbott Laboratories, Cereno Scientific, Gossamer Bio, Respira Therapeutics, and United Therapeutics; V. Franco: Ohio State University receives research support from Abbott Laboratories, Acceleron Pharma, Cordella (Endotronix), Respira Therapeutics, and United Therapeutics; M. A. Aras: None; L. Spikes: Principal investigator for trials sponsored by Acceleron Pharma, Bayer, Gossamer Bio, Insmed, Merck & Co, Respira Therapeutics, and United Therapeutics; D. Grinnan: Received grant support from and is on the grant review committee at Bayer, and received grant support from Respira Therapeutics; C. Satler: Current employee of Respira Therapeutics
- This study is funded by Respira Therapeutics, Inc. Medical writing assistance was provided by Elise Laming, PhD, of ProScribe – Envision Pharma Group, and was funded by Respira Therapeutics, Inc.

Poster presented at Pulmonary Hypertension Association (PHA 2022); June 9–12, 2022; Atlanta, GA, USA

For more information, contact Dr Carol Satler (CSatler@respiratherapeutics.com)